112

Results of a 4-Year Viltolarsen Extension Study of Functional and Safety Outcomes

Edward C. Smith, MD[1]; Paula R. Clemens, MD[2,3]; Anne M. Connolly, MD[4]; Amy D. Harper, MD[5]; Jean K. Mah, MD[6]; Craig M. McDonald, MD[7]; Vamshi K. Rao, MD[8]; Craig M. Zaidman, MD[9]; Robert A. Crozier, PhD[10]; the CINRG DNHS Investigators; Eric P. Hoffman, PhD[11]

[1]Department of Pediatrics, Duke University Medical Center, Durham, North Carolina; [2]Department of Neurology, University of Pittsburgh School of Medicine, Pittsburgh, Pennsylvania; [3]Department of Veterans Affairs Medical Center, Pittsburgh, Pennsylvania; [4]Division of Neurology, Nationwide Children's Hospital, The Ohio State University College of Medicine, Columbus, Ohio; [5]Children's Hospital of Richmond at Virginia Commonwealth University, Richmond, Virginia; [6]Department of Pediatrics, University of Calgary, Calgary, Alberta, Canada; [7]Department of Physical Medicine and Rehabilitation, Department of Pediatrics, UC Davis Health, University of California, Davis, Sacramento, California; [8]Division of Neurology, Ann and Robert H. Lurie Children's Hospital of Chicago, Chicago, Illinois; [9]Department of Neurology, Washington University at St Louis, St Louis, Missouri; [10]NS Pharma, Inc., Paramus, NJ [11]Department of Pharmaceutical Sciences, State University of New York at Binghamton, Binghamton, New York.

INTRODUCTION

- Approximately 8%–10% of patients with Duchenne Muscular Dystrophy (DMD) are amenable to exon 53 skipping treatment, resulting in the production of an internally shortened dystrophin protein containing essential functional portions.[1,2]
- Viltolarsen (VILTEPSO®) is an antisense oligonucleotide designed to treat DMD in patients with a confirmed mutation of the DMD gene amenable to exon 53 skipping. Viltolarsen binds to exon 53 of dystrophin pre-mRNA, resulting in exclusion of this exon during mRNA processing.[3–5]
- In an initial North American phase 2 study (NCT02740972), treatment with viltolarsen in participants with DMD amenable to exon 53 skipping resulted in significant increases in dystrophin production (~6% of normal) at a weekly dose of 80 mg/kg, as assessed by western blot after 24 weeks of treatment.[3] These results were supported by mass spectroscopy.
 - Timed function tests provided evidence of treatment-related clinical benefit when compared with an age-matched and glucocorticoid treatment-matched DMD natural history control group from the Cooperative International Neuromuscular Research Group (CINRG) Duchenne Natural History Study (DNHS).
 - Significant improvements in timed function tests from baseline, including time to stand from supine (TTSTAND), were shown for the viltolarsen-treated group in the phase 2 study.[3]
 - Adverse events (AEs) were classified as mild to moderate with no treatment-related serious AEs reported, and no participants discontinued from the study.[3]
- At the conclusion of the initial study, all participants chose to enroll in a long-term extension (LTE) study (NCT03167255) for up to an additional 192 weeks. An interim analysis at 109 weeks (~2 years) demonstrated that participants who received viltolarsen displayed stabilization in timed function tests compared with the CINRG DNHS historical control group which showed a decline over this time, and similar safety to that observed in the 24-week study.[6]
- Presented here are completed extension study results of >4 years of functional outcomes in viltolarsen-treated patients compared to a matched historical control group. These results show clinically meaningful outcomes from the longest exon 53 skipping study to date.

OBJECTIVE OF THE ANALYSIS

- To present the efficacy and safety of viltolarsen over an additional 192 weeks (~4 years in total) in participants with DMD who are amenable to exon 53 skipping.

METHODS

- This phase 2, multicenter, open-label, extension study of viltolarsen evaluated 16 participants who were originally enrolled in the 24-week study (**Figure 1**).
- Participants were required to remain on a stable dose of glucocorticoids for the duration of the study.
- No participants were excluded from this study because there were no treatment-related serious or severe AEs in the parent study, no patient received treatment that was made for the purpose of dystrophin-related protein induction, and none took any other investigational drugs after the parent study.
- Efficacy assessments were compared to historical controls (CINRG DNHS), who were group-matched for geographic location, age, glucocorticoid use, and ambulatory ability, defined as being able to execute TTSTAND, time to run/walk 10 meters (TTRW), and time to climb 4 stairs (TTCLIMB), at baseline.
 - Patients amenable to exon 44 skipping and those with deletions of exons 1–8 inclusive were excluded from the CINRG DNHS cohort
 - The last week for which efficacy assessments were available for both the viltolarsen and CINRG DNHS groups was week 205; therefore, week 205 is the last assessment date reported.
- Efficacy was measured every 12 weeks by timed function and strength tests, including TTSTAND, TTRW, and TTCLIMB.
- Safety was assessed throughout the study.

Figure 1. Study Design

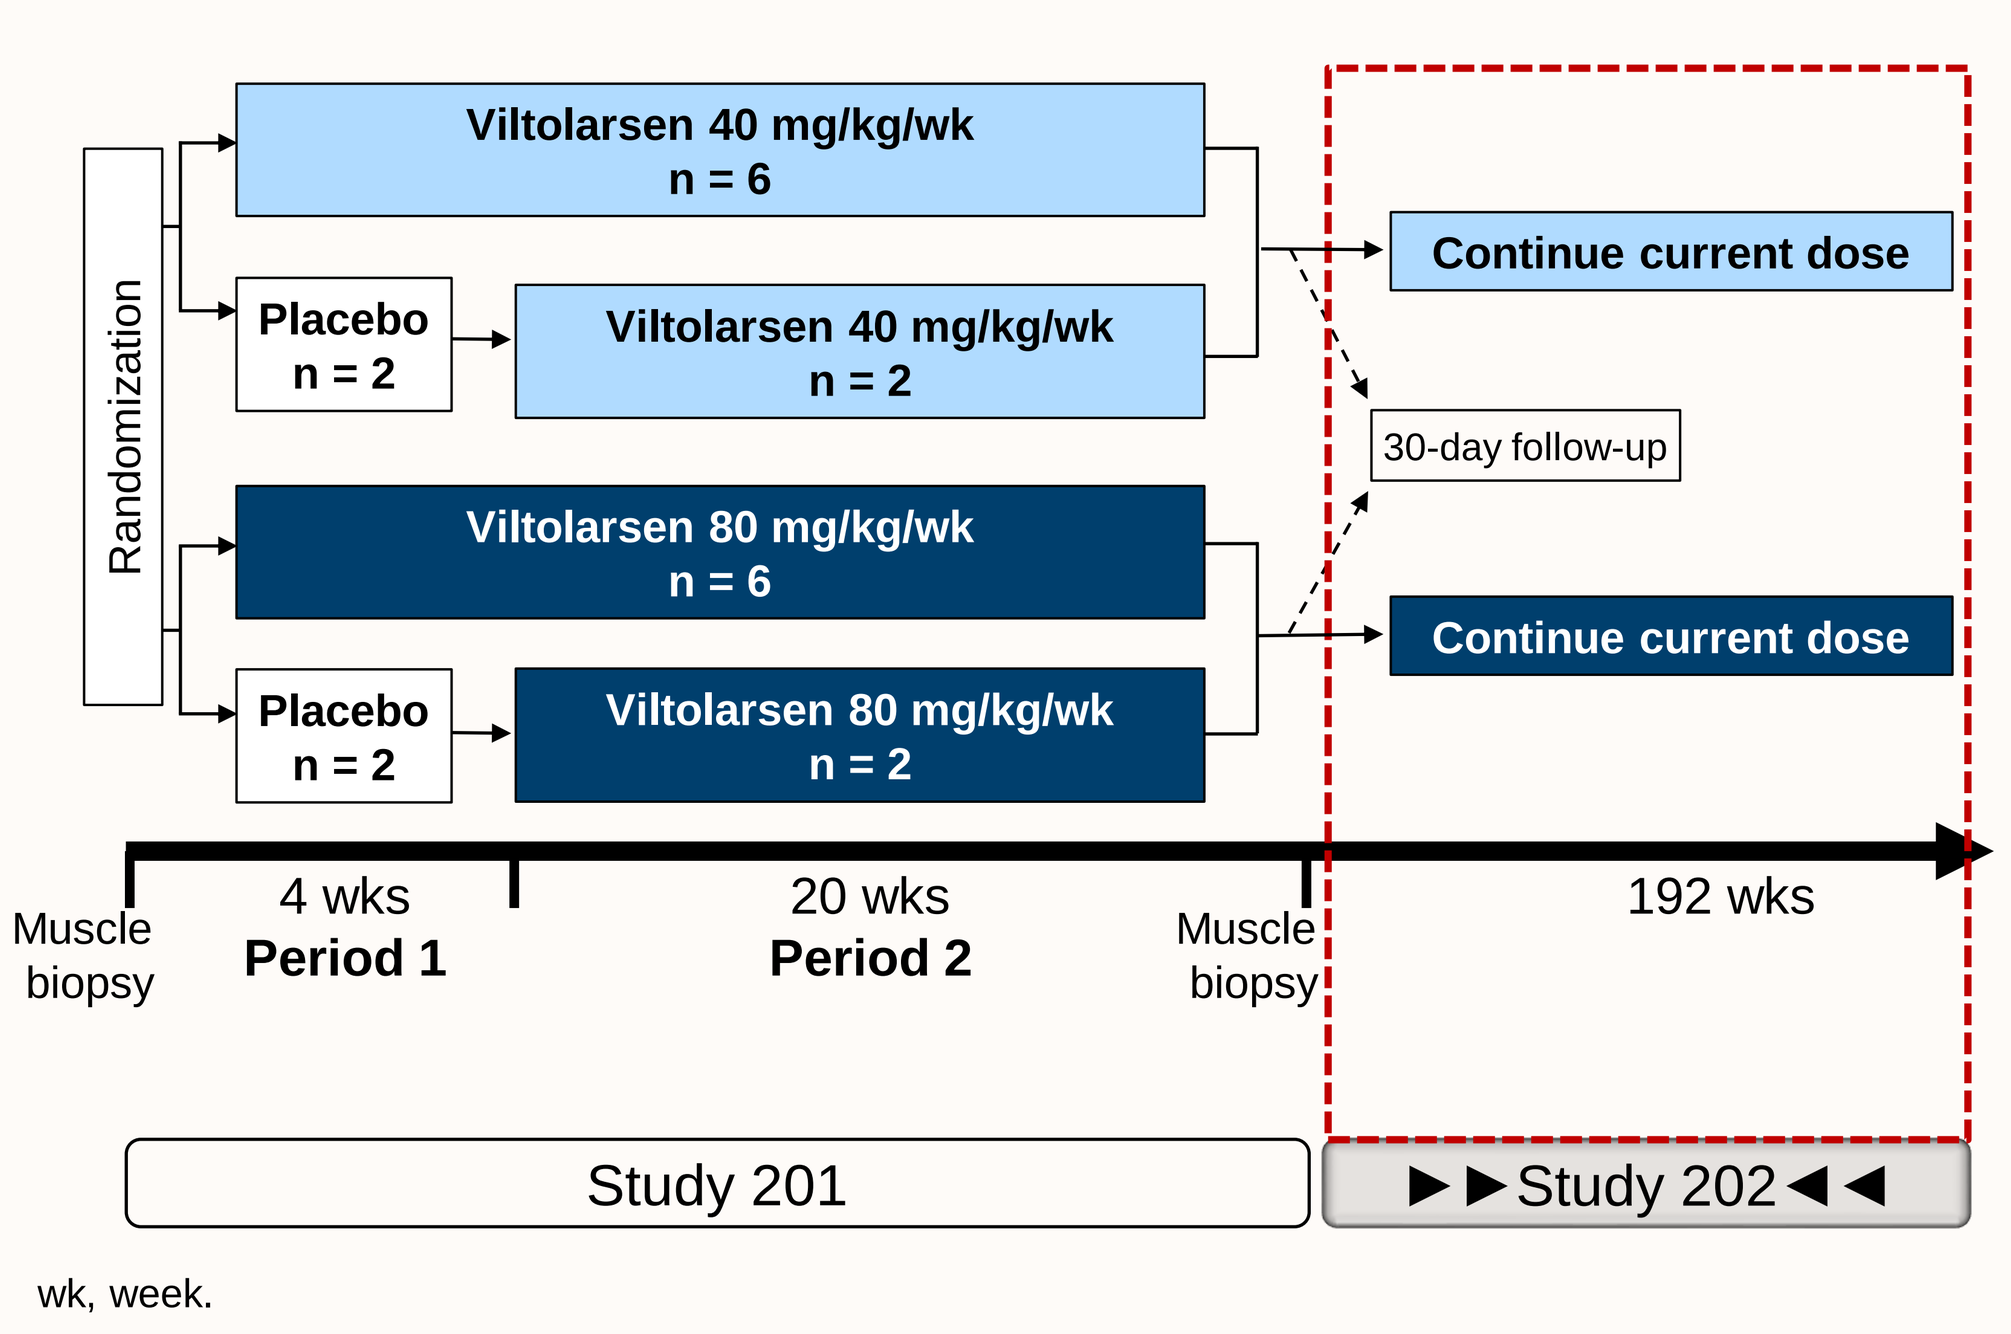
Randomization
Viltolarsen 40 mg/kg/wk
n = 6
Placebo
n = 2
Viltolarsen 40 mg/kg/wk
n = 2
Viltolarsen 80 mg/kg/wk
n = 6
Placebo
n = 2
Viltolarsen 80 mg/kg/wk
n = 2
Continue current dose
30-day follow-up
Continue current dose
Muscle biopsy
4 wks
Period 1
20 wks
Period 2
Muscle biopsy
192 wks
Study 201
►►Study 202◄◄

wk, week.

- Study 201 (**Figure 1**)
 - Boys, aged 4 to <10 years (N = 16), with DMD were enrolled in a randomized, double-blind, placebo-controlled, 4-week safety period of once-weekly infusion of 40 or 80 mg/kg viltolarsen. This was followed by a 20-week open-label period to assess the efficacy and safety of viltolarsen.[3]
- Study 202
 - After completion of the 24-week study, all patients elected to enroll in an additional 192-week treatment period (216 weeks total). Efficacy assessments were conducted every 12 weeks.[6]

RESULTS

Table 1. Baseline Demographics

Characteristics	Viltolarsen			CINRG DNHS control cohort[a]		
	40 mg/kg/wk (n = 8)	80 mg/kg/wk (n = 8)	Total (N = 16)	Exon 53-amenable controls (n = 9)	Non–exon 53-amenable controls (n = 56)	Total (N = 65)
Age, years	7.5 (4.3, 9.8)	7.2 (4.8, 9.8)	7.4 (4.8, 9.8)	6.3 (4.5, 7.8)	7.2 (4.2, 9.6)	7.1 (4.2, 9.6)
Race, n (%)						
White	8 (100)	7 (88)	15 (94)	7 (78)	48 (86)	55 (85)
Black	0	0	0	0	1 (2)	1 (2)
Asian	0	1 (13)	1 (6)	1 (11)	3 (5)	4 (6)
Other	0	0	0	1 (11)	4 (7)	5 (8)
Weight, kg	23.7 (14.9, 30.4)	22.3 (15.5, 35.4)	23.0 (14.9, 35.4)	21.6 (16.6, 28.1)	24.4 (14.8, 38.7)	24.0 (14.8, 38.7)
Height, cm	114.6 (102.5, 123.4)	112.2 (99.4, 127.1)	113.4 (99.4, 127.1)	111.3 (102.2, 122.2)	117.0 (96.1, 135.9)	116.2 (96.1, 135.9)
BMI, kg/m²	17.9 (14.2, 20.0)	17.4 (15.4, 21.9)	17.7 (14.2, 21.9)	17.3 (15.5, 21.6)	17.6 (13.9, 22.9)	17.5 (13.9, 22.9)

All data are mean (range) unless otherwise specified.
[a]See Methods for additional information.
BMI, body mass index; CINRG, The Cooperative International Neuromuscular Research Group; DNHS, Duchenne Natural History Study; wk, week.

- Overall, baseline characteristics between participants in the 2 viltolarsen dosage cohorts were balanced and similar to those of the CINRG DNHS controls (**Table 1**).

Figure 2. TTSTAND: Viltolarsen Cohort vs Natural History Controls

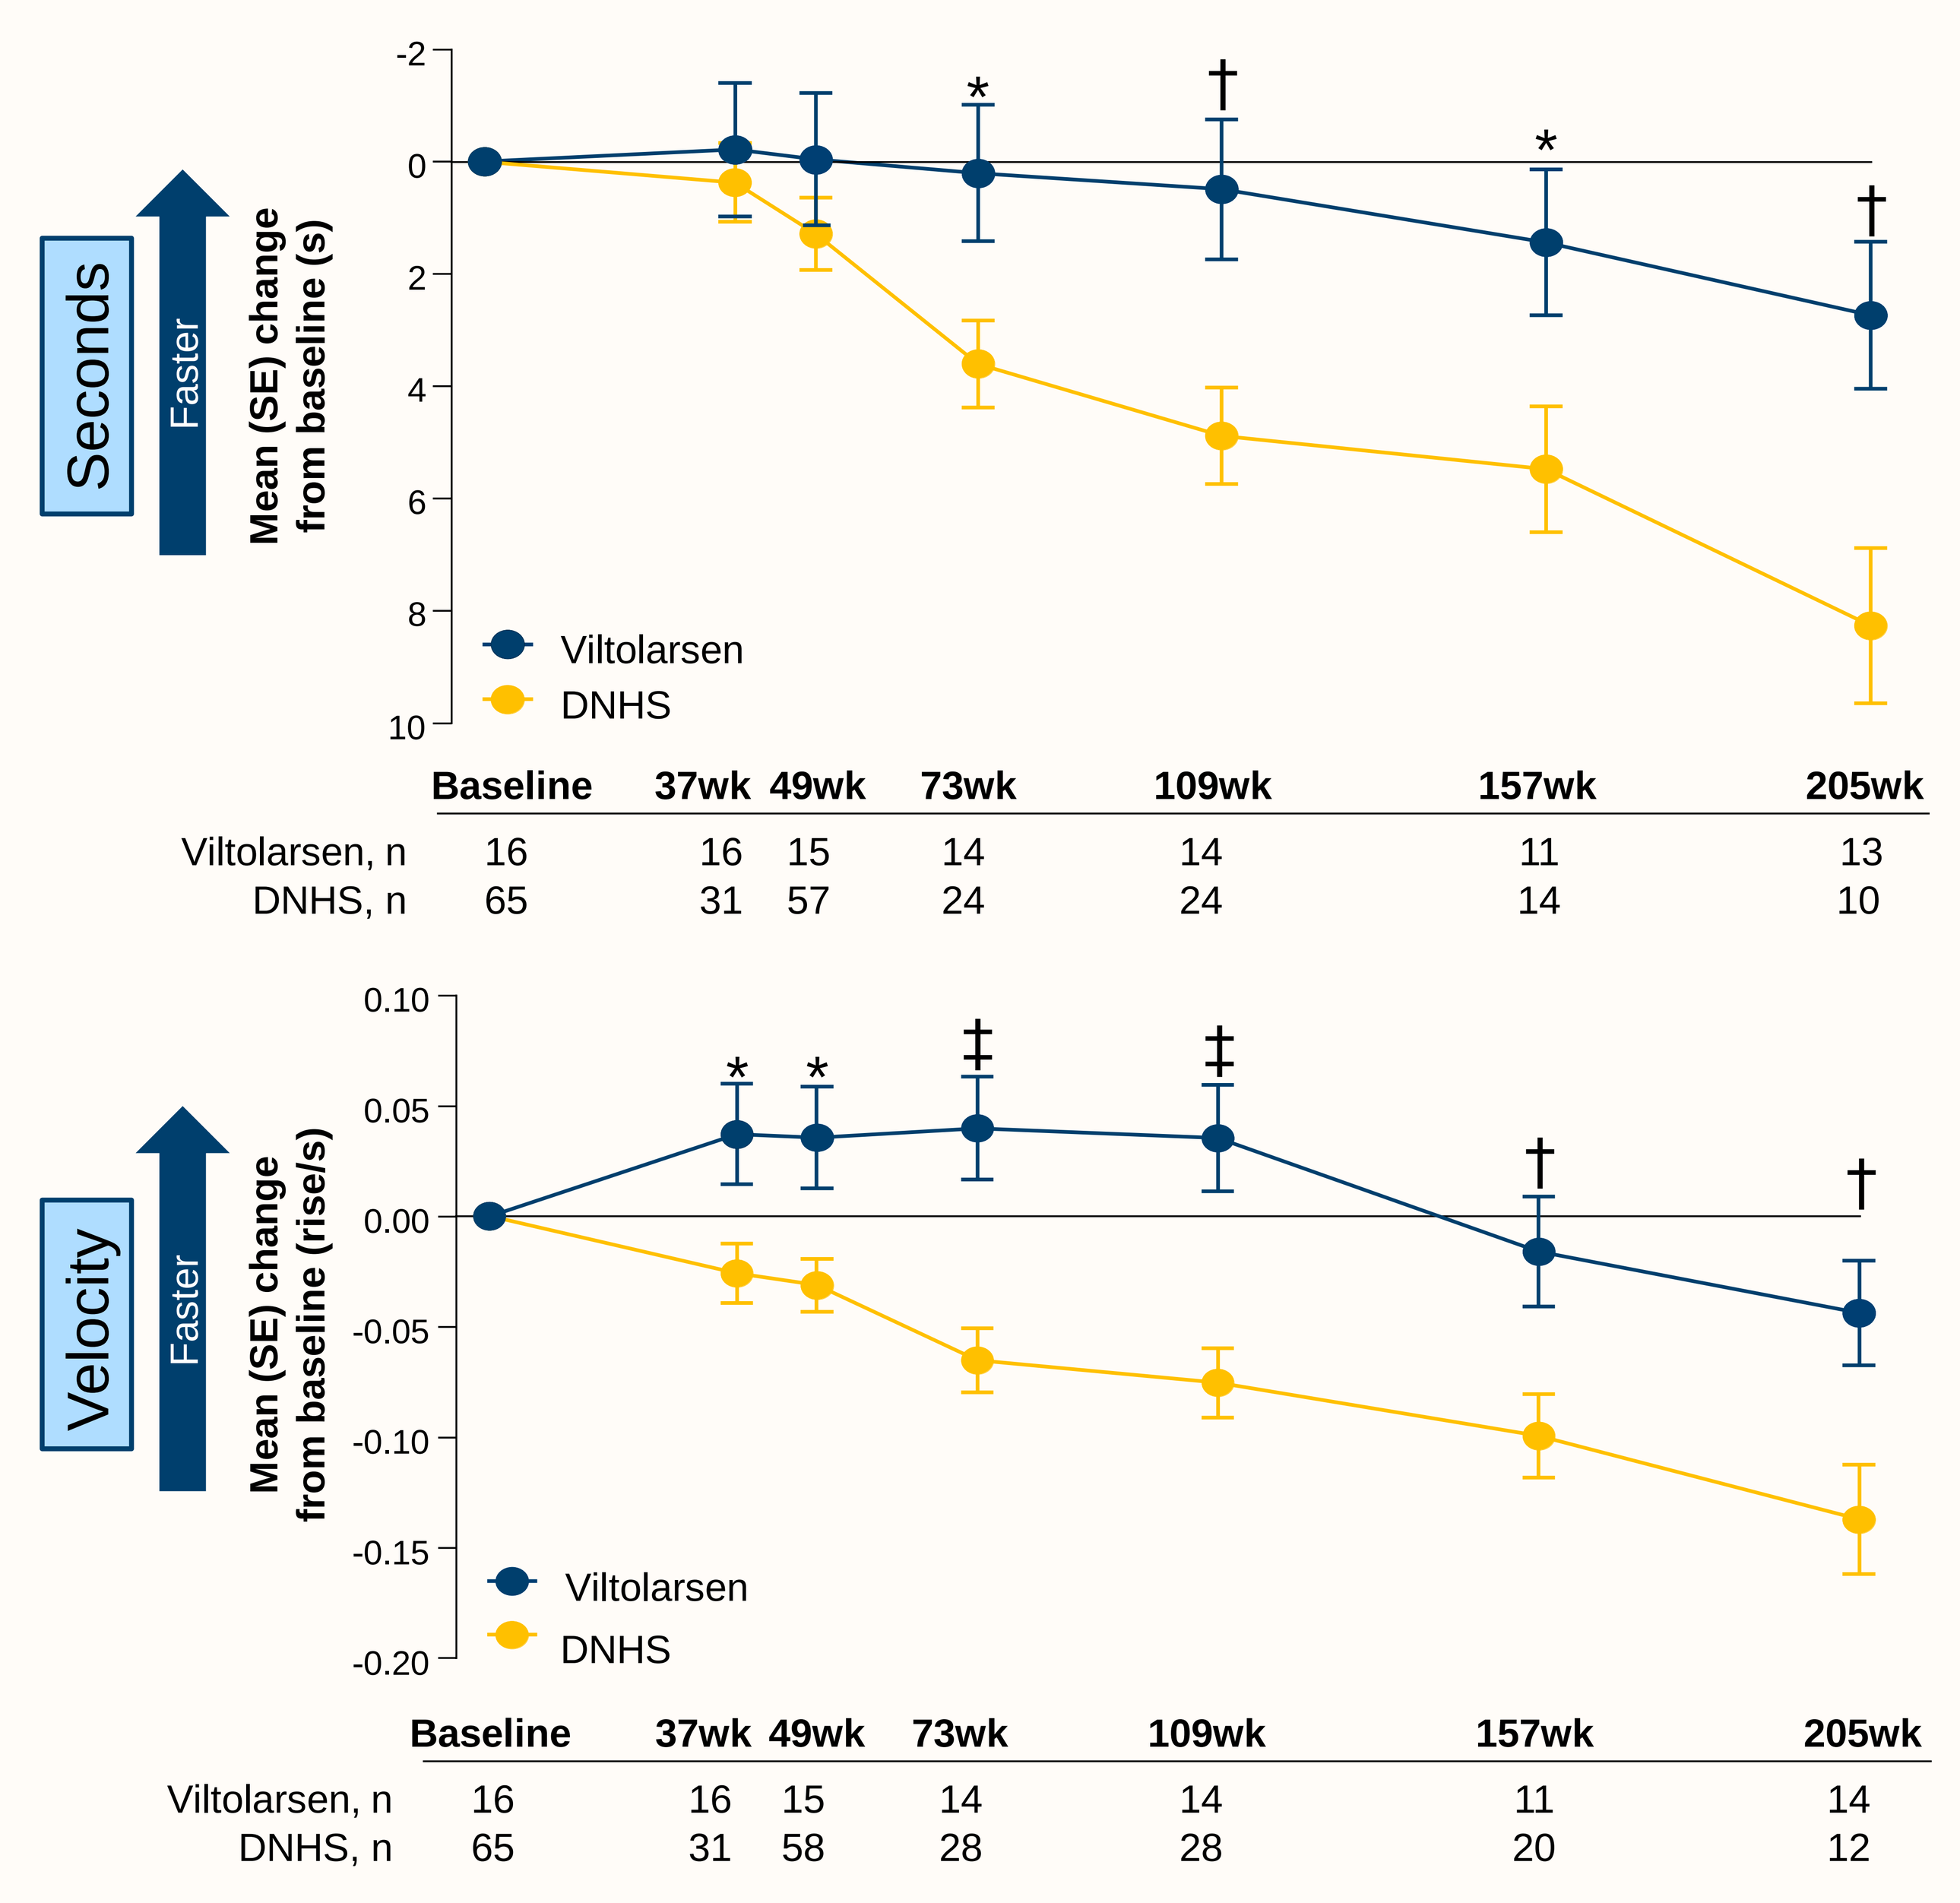
Seconds
Faster
Mean (SE) change from baseline (s)
Viltolarsen
DNHS
Baseline 37wk 49wk 73wk 109wk 157wk 205wk
Viltolarsen, n 16 16 15 14 14 11 13
DNHS, n 65 31 57 24 24 14 10
Velocity
Faster
Mean (SE) change from baseline (rise/s)
Viltolarsen
DNHS
Baseline 37wk 49wk 73wk 109wk 157wk 205wk
Viltolarsen, n 16 16 15 14 14 11 14
DNHS, n 65 31 58 28 28 20 12

*P <0.05; †P <0.01; ‡P ≤0.001.
DNHS, Duchenne Natural History Study; SE, standard error; TTSTAND, time to stand from supine; wk, week.

- Participants who received viltolarsen showed maintenance of motor function over the first 2 years treatment, and significant slowing of disease progression over the following 2 years, whereas the CINRG DNHS controls experienced functional decline (**Figure 2**).
- Beginning at week 73 and through week 205, TTSTAND seconds and velocity were statistically significantly different for participants who received viltolarsen compared with CINRG DNHS controls (velocity was also different between groups as early as week 37).

Figure 3. TTRW: Viltolarsen Cohort vs Natural History Controls

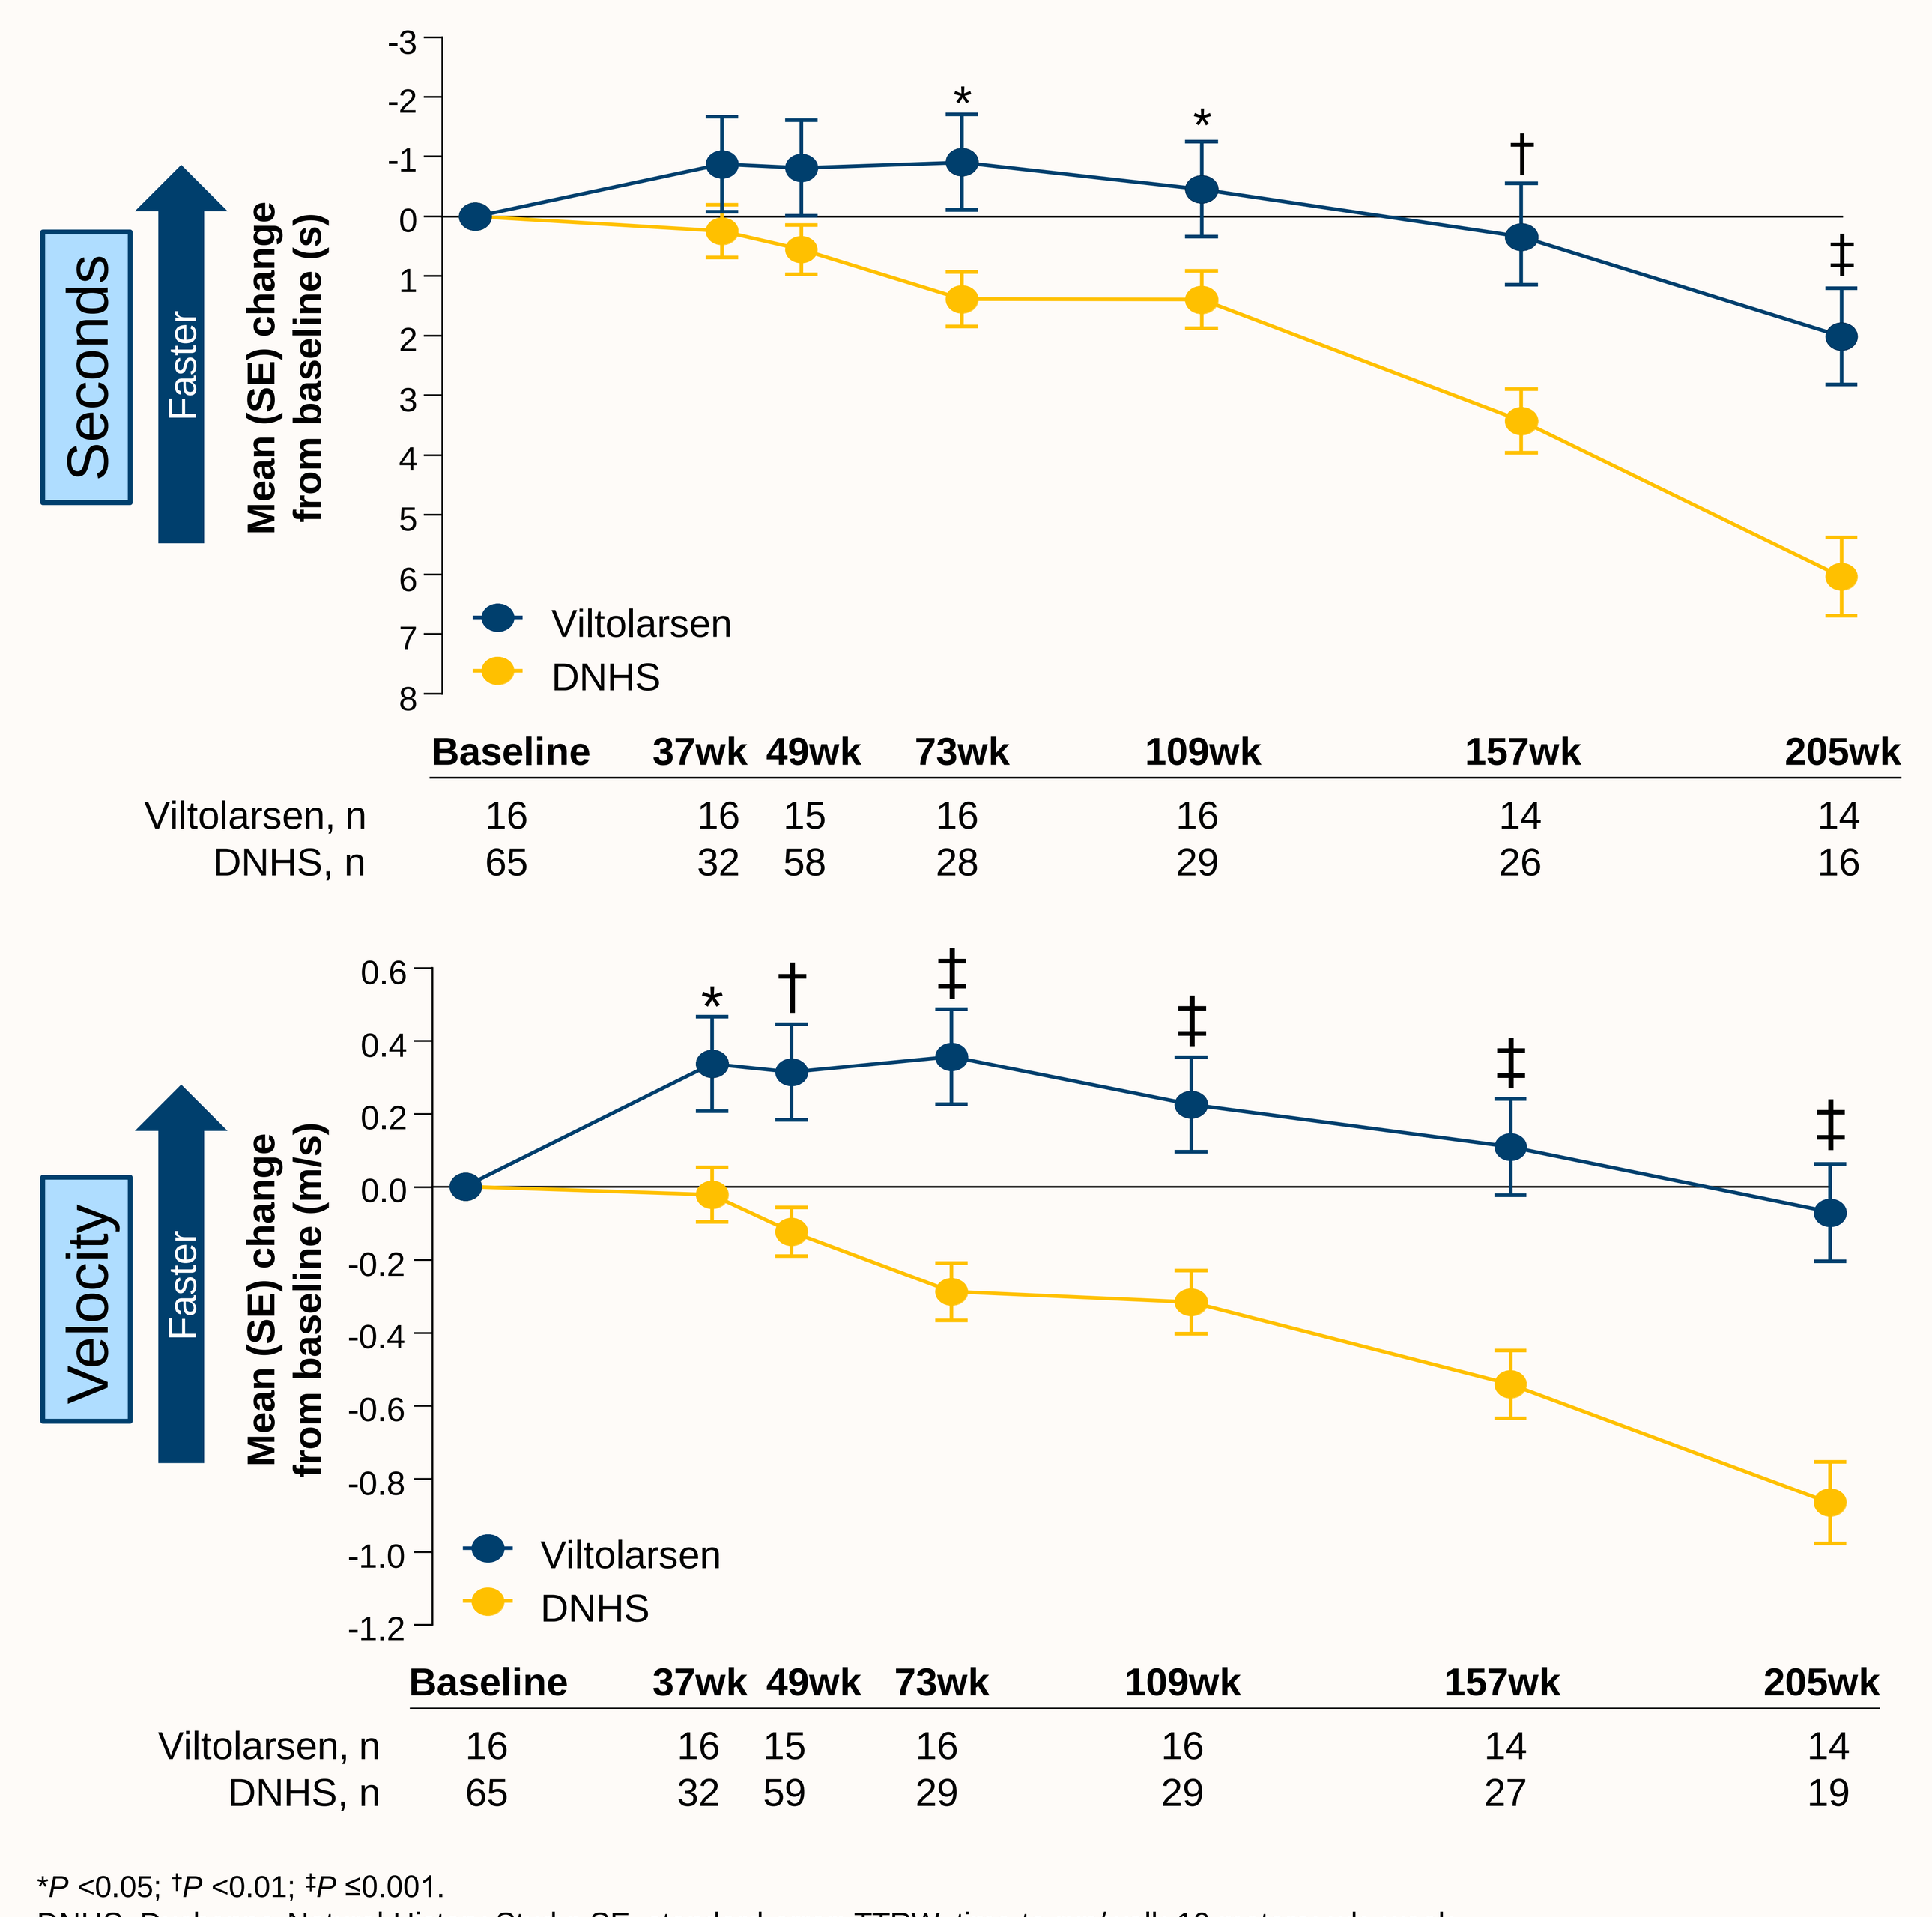
Seconds
Faster
Mean (SE) change from baseline (s)
Viltolarsen
DNHS
Baseline 37wk 49wk 73wk 109wk 157wk 205wk
Viltolarsen, n 16 16 15 16 16 14 14
DNHS, n 65 32 58 28 29 26 16
Velocity
Faster
Mean (SE) change from baseline (m/s)
Viltolarsen
DNHS
Baseline 37wk 49wk 73wk 109wk 157wk 205wk
Viltolarsen, n 16 16 15 16 16 14 14
DNHS, n 65 32 59 29 29 27 19

*P <0.05; †P <0.01; ‡P ≤0.001.
DNHS, Duchenne Natural History Study; SE, standard error; TTRW, time to run/walk 10 meters; wk, week.

- Change from baseline for TTRW showed maintenance of motor function over the first 2 years and significant slowing of disease progression over the following 2 years for viltolarsen-treated participants vs CINRG DNHS controls (**Figure 3**).
- Beginning at week 73 and through week 205, TTWR seconds and velocity were statistically significantly different for participants who received viltolarsen compared with CINRG DNHS controls (velocity was also different between groups as early as week 37).

Figure 4. TTCLIMB: Viltolarsen Cohort vs Natural History Controls

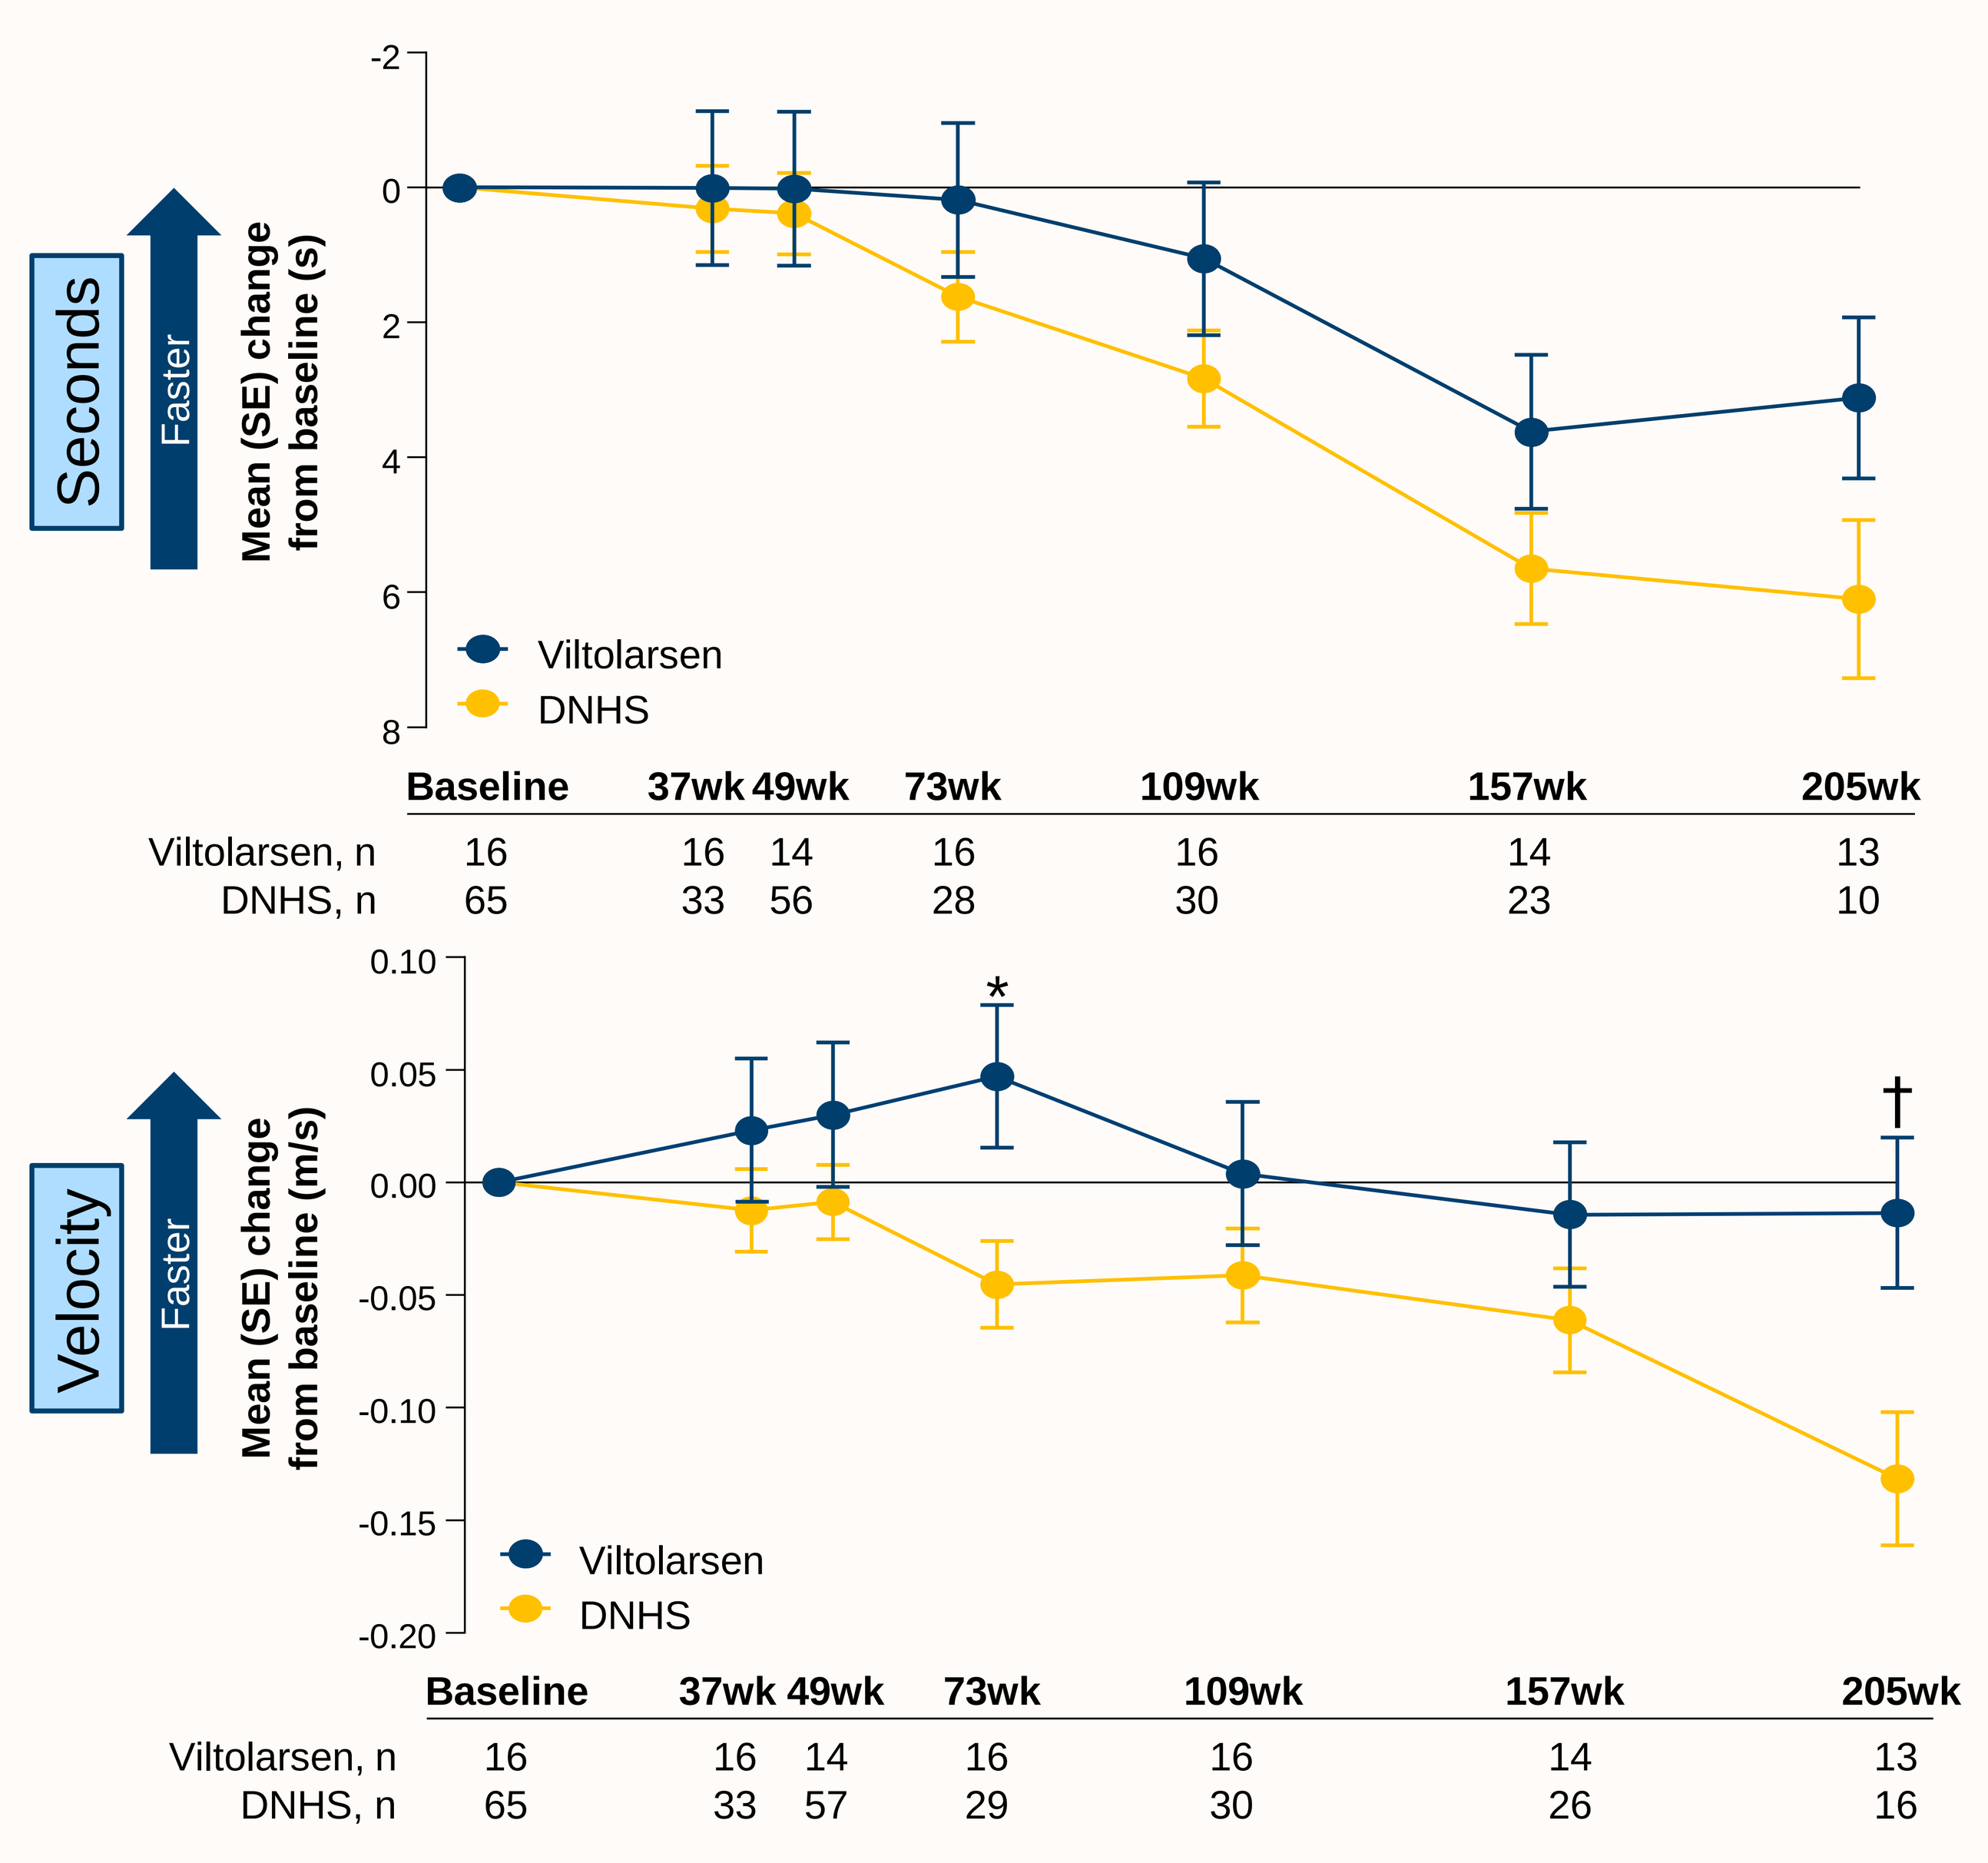
Seconds
Faster
Mean (SE) change from baseline (s)
Viltolarsen
DNHS
Baseline 37wk 49wk 73wk 109wk 157wk 205wk
Viltolarsen, n 16 16 14 16 16 14 13
DNHS, n 65 33 56 28 30 23 10
Velocity
Faster
Mean (SE) change from baseline (m/s)
Viltolarsen
DNHS
Baseline 37wk 49wk 73wk 109wk 157wk 205wk
Viltolarsen, n 16 16 14 16 16 14 13
DNHS, n 65 33 57 29 30 26 16

*P <0.05; †P <0.01.
DNHS, Duchenne Natural History Study; SE, standard error; TTCLIMB, time to climb 4 stairs; wk, week.

- TTCLIMB was numerically equivalent or improved at all time points in participants treated with viltolarsen compared with CINRG DNHS controls, with TTCLIMB velocity being statistically significantly different at weeks 73 and 205 between groups (**Figure 4**).

Table 2. Safety Assessment

	Viltolarsen participants		
Participants with:	40 mg/kg/wk (n = 8)	80 mg/kg/wk (n = 8)	Total (N = 16)
Any TEAE, n (%)	8 (100)	8 (100)	16 (100)
Any drug-related TEAE, n (%)	0	1 (13)	1 (6)
Any serious treatment-related AE, n (%)	0	0	0
Study drug discontinuation due to TEAE, n (%)	0	0	0
Death, n (%)	0	0	0

AE, adverse event; TEAE, treatment-emergent AE; wk, week.

- All 16 participants experienced a treatment-emergent AE (TEAE; **Table 2**).
- No participants experienced any serious treatment-related AEs, discontinued the study drug due to a TEAE, or died.
- Three participants reported a total of 4 serious AEs, none of which was considered related to study medication (broken left tibia and fibula, broken right femur, rhabdomyolysis, and left tibia/fibula fracture).

Table 3. Common TEAEs (Preferred Term in ≥25% of Participants)

	Viltolarsen participants		
TEAEs	40 mg/kg/wk (n = 8)	80 mg/kg/wk (n = 8)	Total (N = 16)
Cough, n (%)	5 (63)	5 (63)	10 (63)
Nasopharyngitis, n (%)	4 (50)	5 (63)	9 (56)
Insect bite, n (%)	4 (50)	2 (25)	6 (38)
Rash, n (%)	2 (25)	4 (50)	6 (38)
Vomiting, n (%)	3 (38)	3 (38)	6 (38)
Fever, n (%)	2 (25)	3 (38)	5 (31)
Fall, n (%)	4 (50)	1 (13)	5 (31)
Headache, n (%)	3 (38)	2 (25)	5 (31)
Nasal congestion, n (%)	3 (38)	2 (25)	5 (31)
Influenza, n (%)	3 (38)	1 (13)	4 (25)

TEAE, treatment-emergent adverse event; wk, week.

- The most frequently reported TEAEs were cough and nasopharyngitis (**Table 3**).

CONCLUSIONS

- Participants treated with viltolarsen in this long-term extension study for up to an additional 192 weeks (up to 216 weeks [4 years]) showed maintenance of function over the first 2 years and significantly delayed disease progression over the following 2 years in timed function tests compared with CINRG DNHS controls, which declined over this same time period.
- Viltolarsen was well tolerated over the duration of this 4-year study, with most reported TEAEs being mild or moderate; no discontinuations or deaths were reported.
- The study outcome of better motor function vs historical controls, and a favorable safety profile, have been demonstrated in this trial, the longest exon 53 skipping therapy trial to date.
- Viltolarsen can be considered as an important part of the treatment strategy for patients with DMD who are amenable to exon 53 skipping.

References

1. Bladen CL, et al. *Hum Mutat*. 2015;36(4):395–402. **2.** Komaki H, et al. *Sci Transl Med.* 2018;10(437). **3.** Clemens P, et al. *JAMA Neurol.* 2020;77(8):982–91. **4.** Watanabe N, et al. *Mol Ther Nucleic Acids.* 2018;13:442–9. **5.** Viltolarsen (VILTEPSO®) injection. NS Pharma; 2000. https://www.viltepso.com/prescribing-information. Accessed February 7, 2023. **6.** Clemens P, et al. *J Neuromuscul Dis*. 2022;9(4):493–501.

Acknowledgments

- The authors thank all study participants and their families, as well as the study team members at participating CINRG sites for assistance in collecting and managing the data.
- Medical writing and editorial assistance were provided by Gregory Suess, PhD, of AlphaBioCom, LLC, under the direction of the authors, and funded by NS Pharma, Inc.
- This study was funded by NS Pharma, Inc.

Presented at the 2023 MDA Clinical & Scientific Conference; 19–22 March 2023; Dallas, TX, USA.

NS Pharma